First results from the phase I trial of the ATR inhibitor, ART0380, in advanced solid tumors

680P

Kathleen Moore,[1] Manish R. Patel,[2] Gerald S. Falchook,[3] Elisa Fontana,[4] Babar Bashir,[5] Cesar A. Perez,[6] Minal Barve,[7] Susanna Ulahannan,[1] Helen Millward,[8] Amelia Fielding,[8] Bryony Harrop,[8] Nicola Little,[8] Tanya Coleman,[8] Alison Wilby,[9] Sara Busacca,[8] Sarah Holt,[8] Suraj Menon,[8] Desirée Headley,[8] Ian Smith,[8] Melissa Johnson[10]

[1]Stephenson Cancer Center at the Univ of Oklahoma, TSET/SCRI Phase 1 Drug Development Unit, Oklahoma City, OK, USA; [2]Florida Cancer Specialists/Sarah Cannon Research Institute, Sarasota, FL, USA; [3]Sarah Cannon Research Institute at HealthONE, Denver, CO, USA; [4]Sarah Cannon Research Institute UK, London, UK; [5]Sarah Cannon Research Institute and Sidney Kimmel Cancer Center, Thomas Jefferson University Hospital, Philadelphia, PA, USA; [6]Sarah Cannon Research Institute at Florida Cancer Specialists, Lake Nona, Orlando, FL. USA; [7]Mary Crowley Cancer Research, Dallas TX, USA; [8]Artios Pharma Ltd, UK; [9]Seda Pharmaceutical Development Services, Manchester, UK; [10]Sarah Cannon Research Institute at Tennessee Oncology, Nashville, TN, USA

BACKGROUND

- Ataxia telangiectasia and Rad-3 related (ATR) kinase is a critical protein involved in the sensing and signaling of replication stress.
- ATR is activated in response to a variety of DNA damaging agents that induce formation of single-strand DNA lesions during S-phase.
- Inhibition of ATR leads to cell cycle abrogation, collapse of stalled replication forks, increased levels of DNA damage, and cell death.[1]

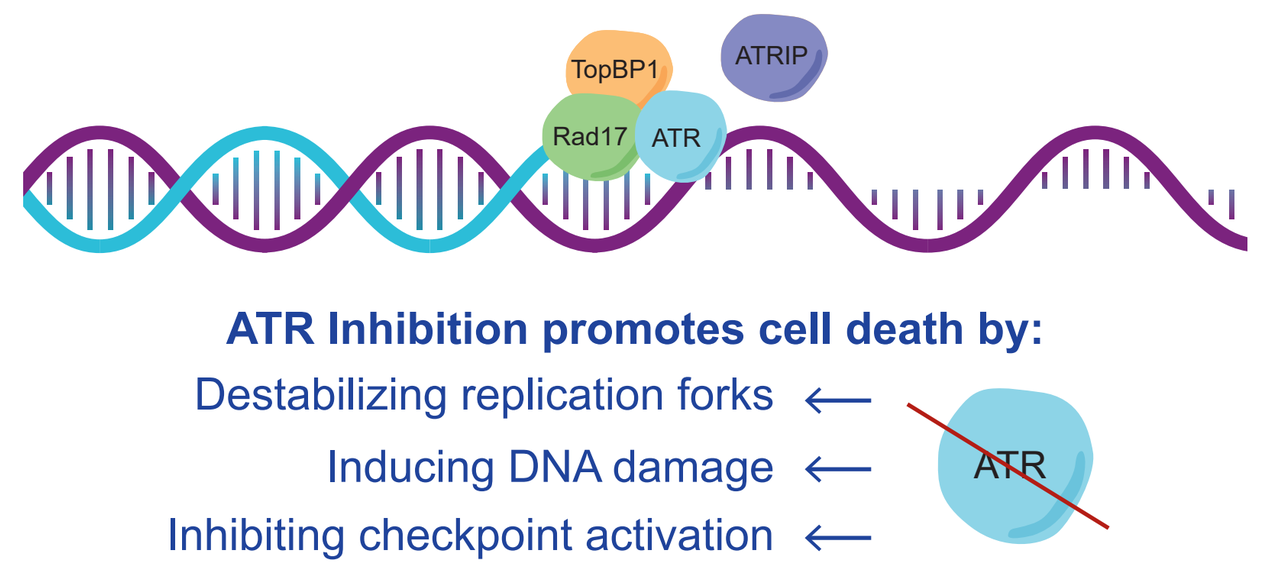
TopBP1
ATRIP
Rad17
ATR
ATR Inhibition promotes cell death by:
Destabilizing replication forks
Inducing DNA damage
Inhibiting checkpoint activation
ATR

METHODS

- The phase 1/1b trial (NCT04657068) is assessing the safety and tolerability, pharmacokinetics, pharmacodynamics, and preliminary efficacy of ART0380.
- Patient population:
 - ≥18 years with advanced or metastatic solid tumors
 - ECOG Performance Status 0–2
 - Hemoglobin ≥9 g/dL, absolute neutrophil count (ANC) ≥1.5 x 10^9/L, platelets ≥100 x 10^9/L.
- Ataxia-telangiectasia-mutated (ATM) deficiency identified by Artios' ATR-ID algorithm (bespoke algorithm to predict ATM protein deficiency using mutational impact) or IHC H-score ≤10, or percentage staining at 0 in ≥90% of tumor cells.
- Cancers with molecular alterations presumed to cause replication stress identified by Artios' DcoDeR platform.

Figure 1: ART0380 dose escalation

- Patients received ART0380 dosed once daily (QD) on a continuous (cont) or intermittent (int) schedule.

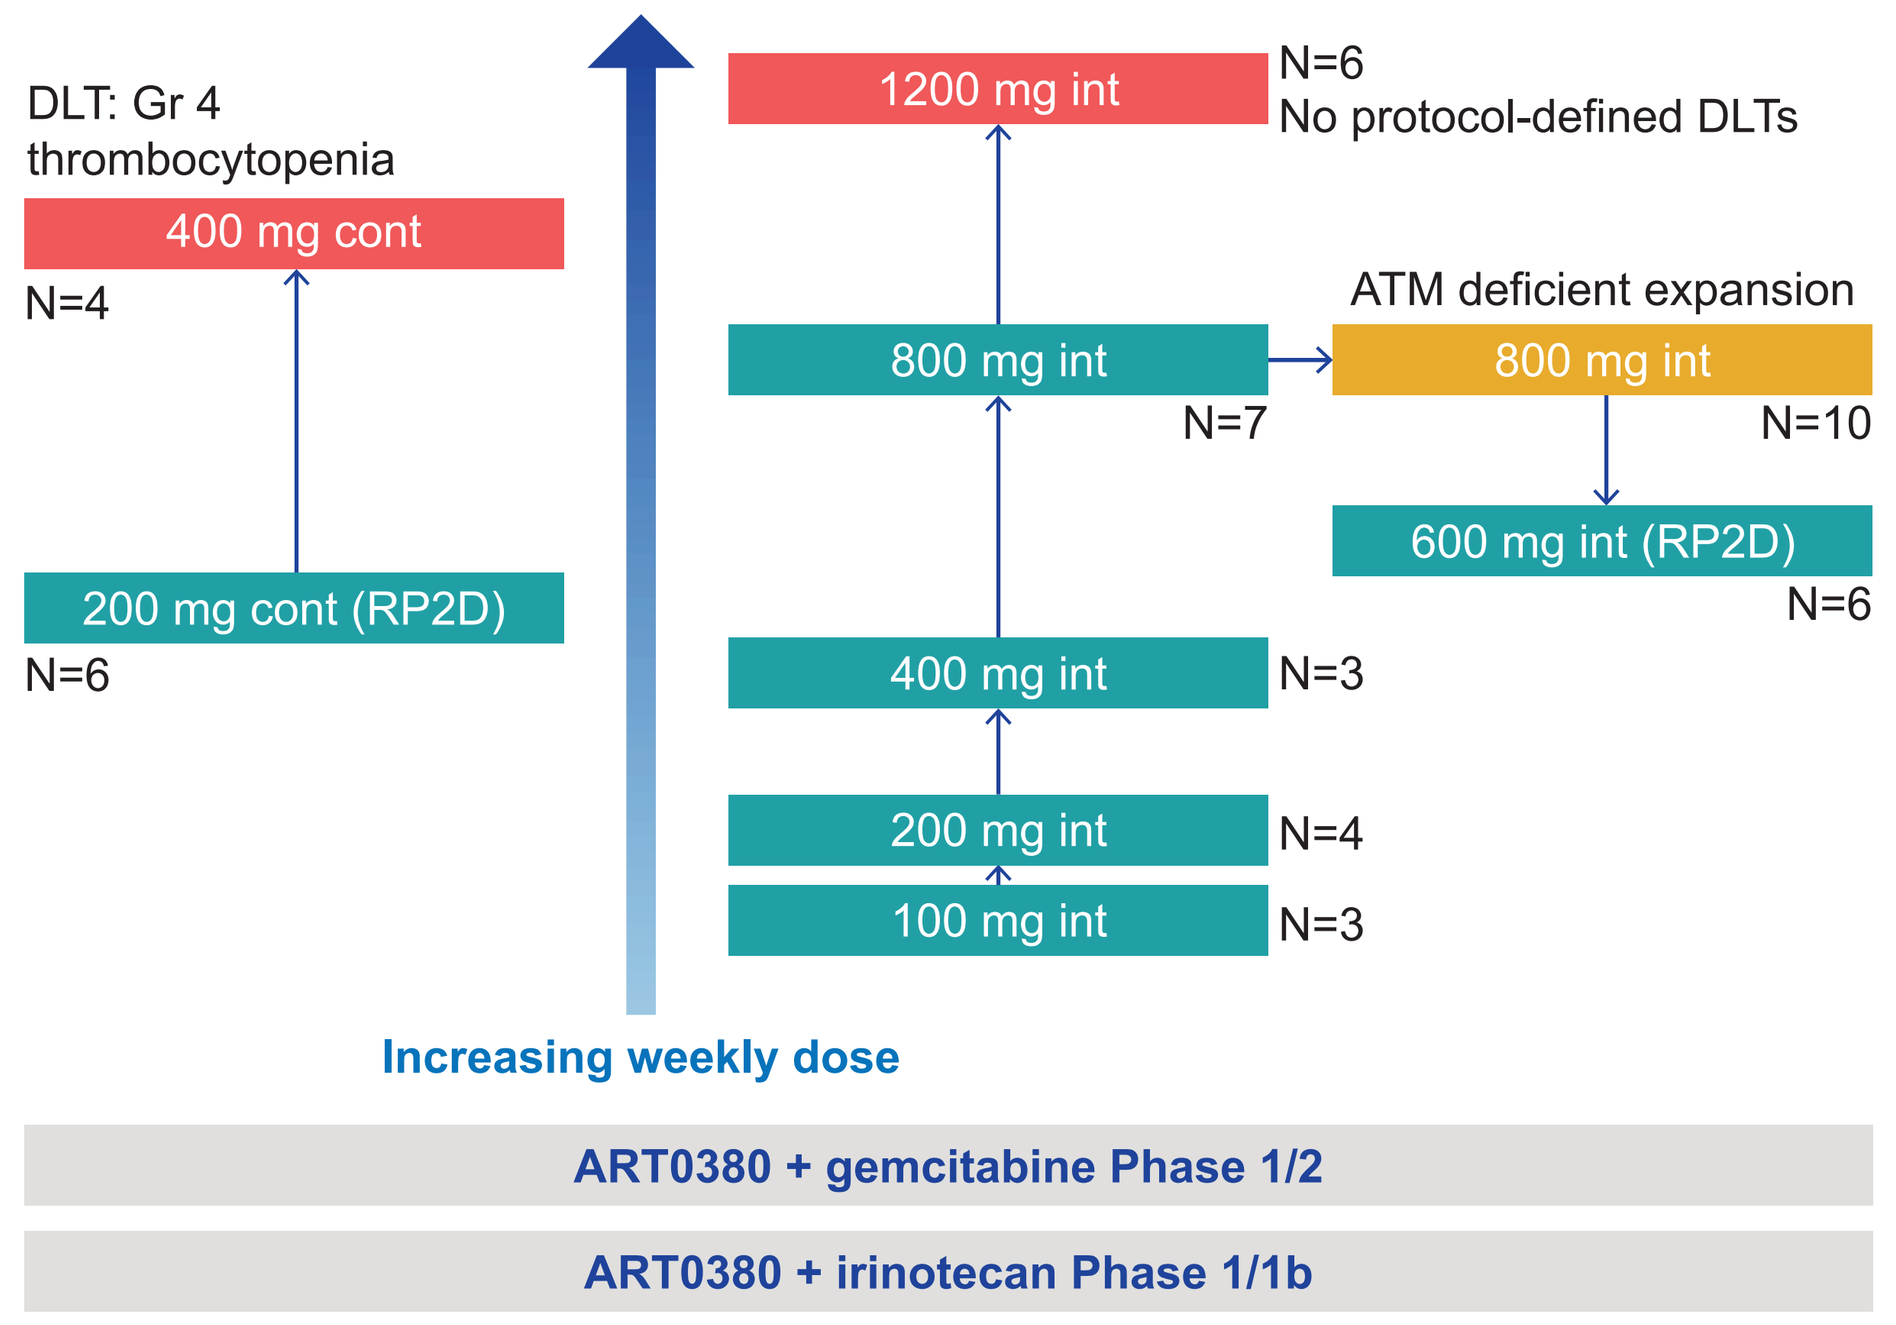
DLT: Gr 4 thrombocytopenia
400 mg cont
N=4
200 mg cont (RP2D)
N=6
1200 mg int
N=6
No protocol-defined DLTs
800 mg int
N=7
ATM deficient expansion
800 mg int
N=10
600 mg int (RP2D)
N=6
400 mg int
N=3
200 mg int
N=4
100 mg int
N=3
Increasing weekly dose
ART0380 + gemcitabine Phase 1/2
ART0380 + irinotecan Phase 1/1b
Tolerated dose
Non-feasible dose
Non-tolerated dose

RESULTS

Patient demographics and disease characteristics

- Baseline demographics and characteristics were available for 49 patients from 8 monotherapy cohorts (Table 1).
- 36 patients with ≥1 post-baseline scan at a biologically effective dose based on target exposure from preclinical efficacy were included in the efficacy analysis set.

Table 1: Baseline demographics and disease characteristics

Parameter	All patients N = 49 (%)
Sex, n (%)	
Male	22 (45)
Female	27 (55)
Age (years), median (range)	62 (30–80)
≥65 years, n (%)	21 (43)
ECOG status, n (%)	
0	21 (43)
1	28 (57)
Lines of prior systemic therapy, n (%)	
≤3	21 (43)
≥4	28 (57)
ATM deficient, n (%)	21 (43)
Prior platinum, n (%)	33 (67)
Prior PARP inhibitor, n (%)	14 (29)
Prior PD-(L)1 inhibitor, n (%)	17 (35)

Tumor types, n (%)	All patients N = 49 (%)
Breast	7 (14)
Colon	6 (12)
Prostate	6 (12)
Other[a]	6 (12)
Ovary	4 (8)
Uterus/endometrial	4 (8)
Pancreas	3 (6)
Head/neck	3 (6)
Rectal	3 (6)
Sarcoma	2 (4)
Esophageal	1 (2)
Lung	1 (2)
Peritoneum	1 (2)
Skin (non-melanoma)	1 (2)
Small intestine	1 (2)

[a]Other: 3 x cholangiocarcinoma, adenoid cystic carcinoma of right maxillary sinus, neuroendocrine, anal.

RESULTS

Figure 2: Pharmacokinetics of ART0380 after multiple dosing

1. Rapidly absorbed; half-life ~8.5 hr with reproducible PK profile (Figure 2A).
2. Linear Cmax and AUC consistent across all doses and schedules (Figure 2B).
3. Low variability leads to predictable patient experience (Figures 2A and 2B).
4. Biologically effective doses: ≥200 mg cont; ≥400 mg int.
5. RP2Ds: 200 mg cont and 600 mg int (Figure 2C).

A) Plasma concentration

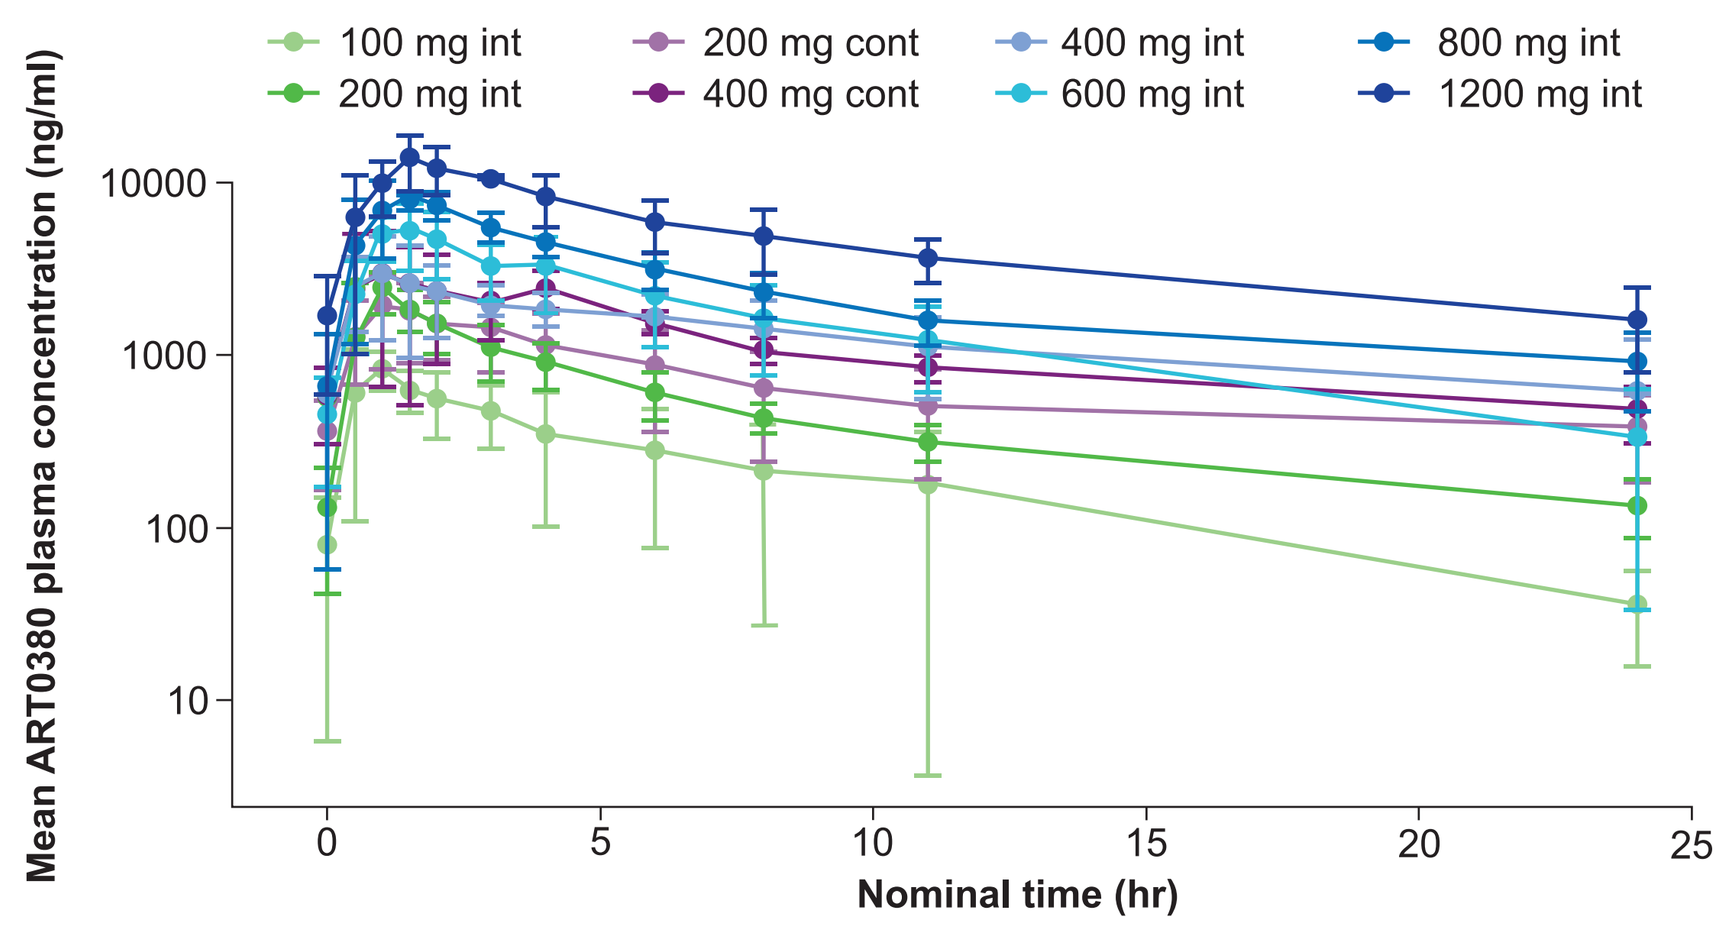
100 mg int
200 mg int
200 mg cont
400 mg cont
400 mg int
600 mg int
800 mg int
1200 mg int
Mean ART0380 plasma concentration (ng/ml)
10000
1000
100
10
0
5
10
15
20
25
Nominal time (hr)

B) Dose-normalized Cmax and AUC0–24

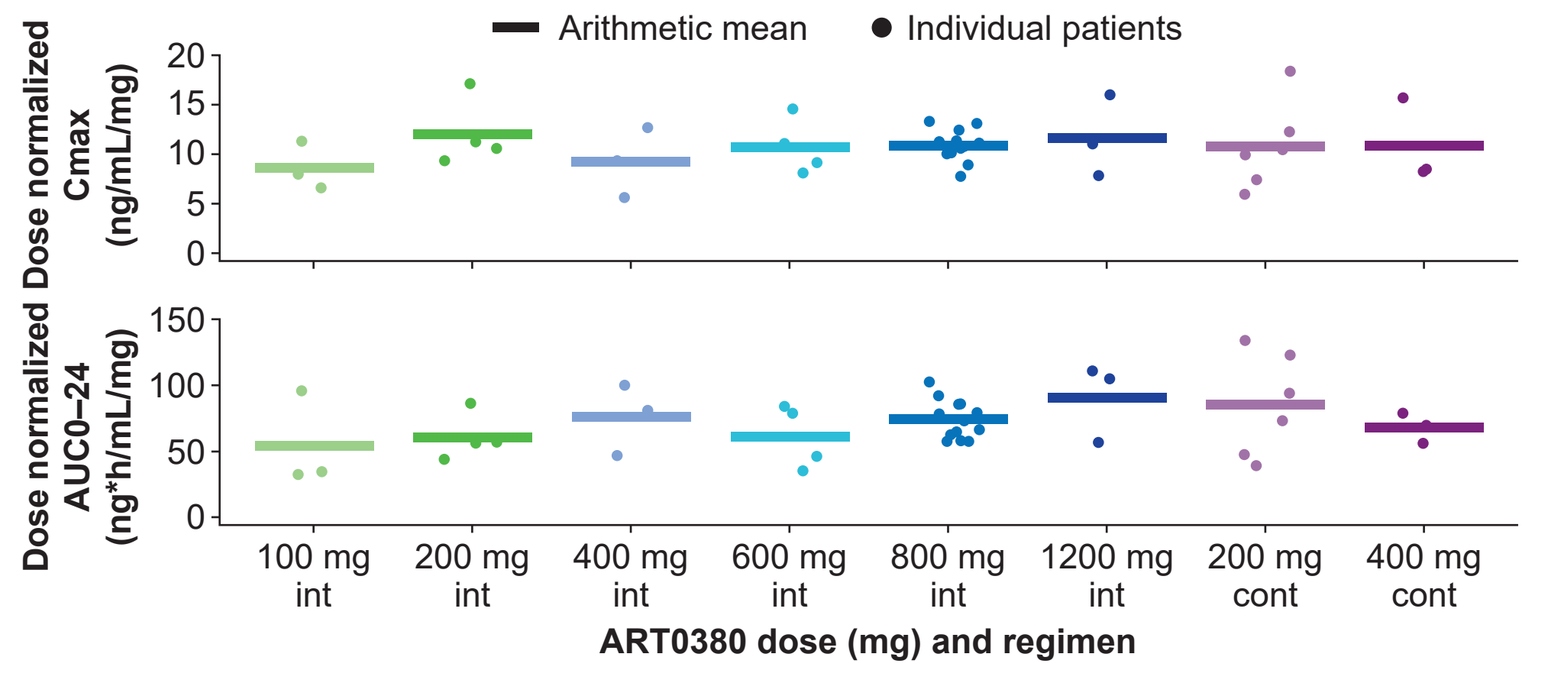
Arithmetic mean
Individual patients
Dose normalized Cmax (ng/mL/mg)
Dose normalized AUC0–24 (ng*h/mL/mg)
100 mg int
200 mg int
400 mg int
600 mg int
800 mg int
1200 mg int
200 mg cont
400 mg cont
ART0380 dose (mg) and regimen

C) Plasma concentration at RP2D

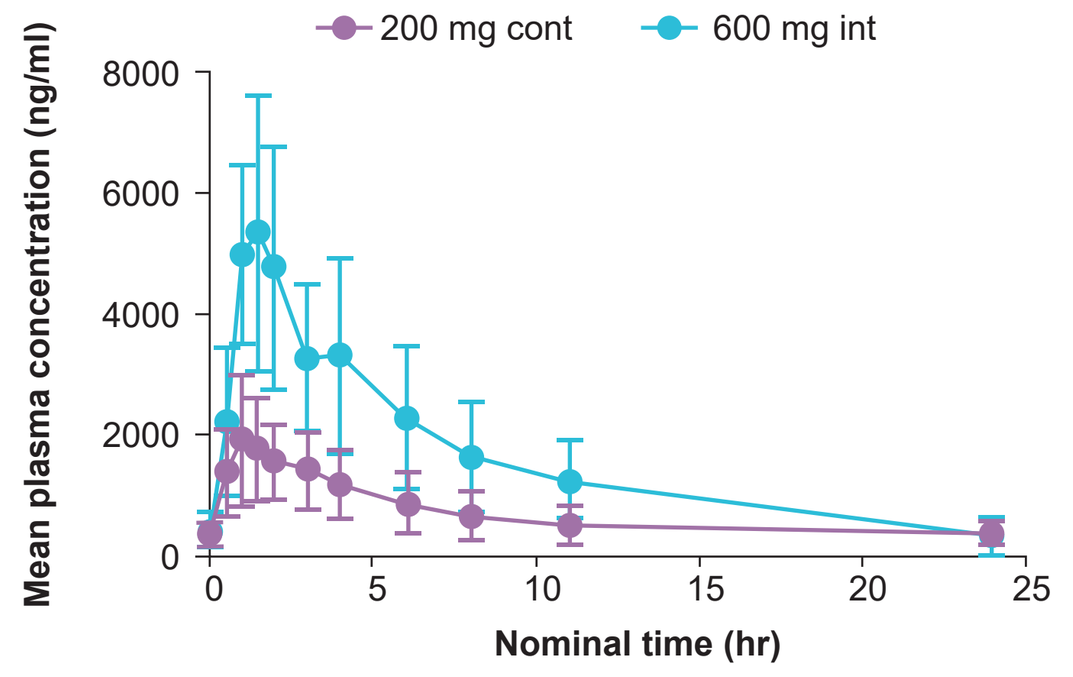
200 mg cont
600 mg int
Mean plasma concentration (ng/ml)
8000
6000
4000
2000
0
Nominal time (hr)

Figure 3: Percentage of γH2Ax expression in CTCs before and after int dosing

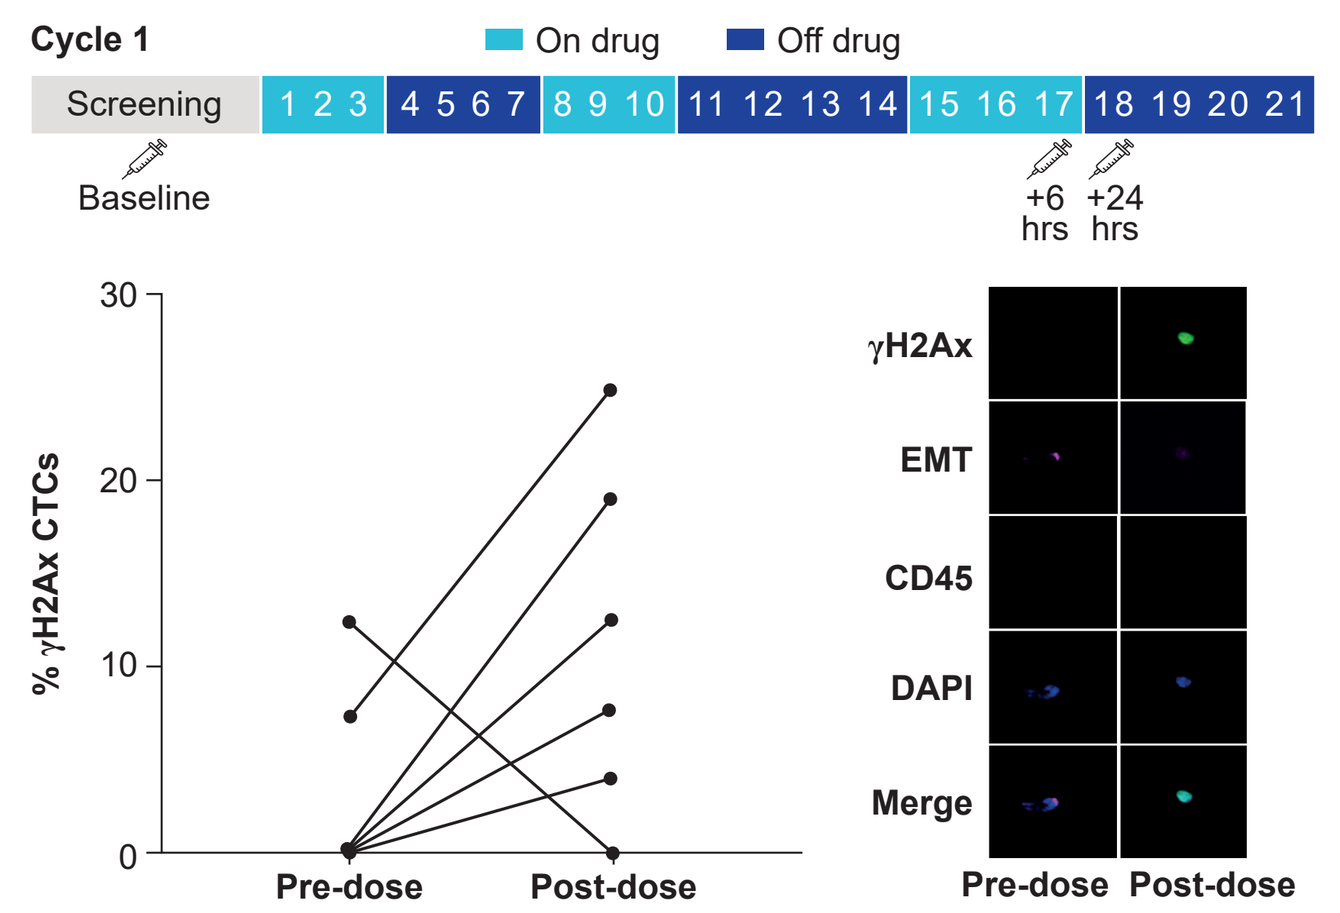
Cycle 1
On drug
Off drug
Screening
1 2 3
4 5 6 7
8 9 10
11 12 13 14
15 16 17
18 19 20 21
Baseline
+6 hrs
+24 hrs
% γH2Ax CTCs
Pre-dose
Post-dose
γH2Ax
EMT
CD45
DAPI
Merge
Pre-dose
Post-dose

Figure 4: PFS with or without a ctDNA molecular response

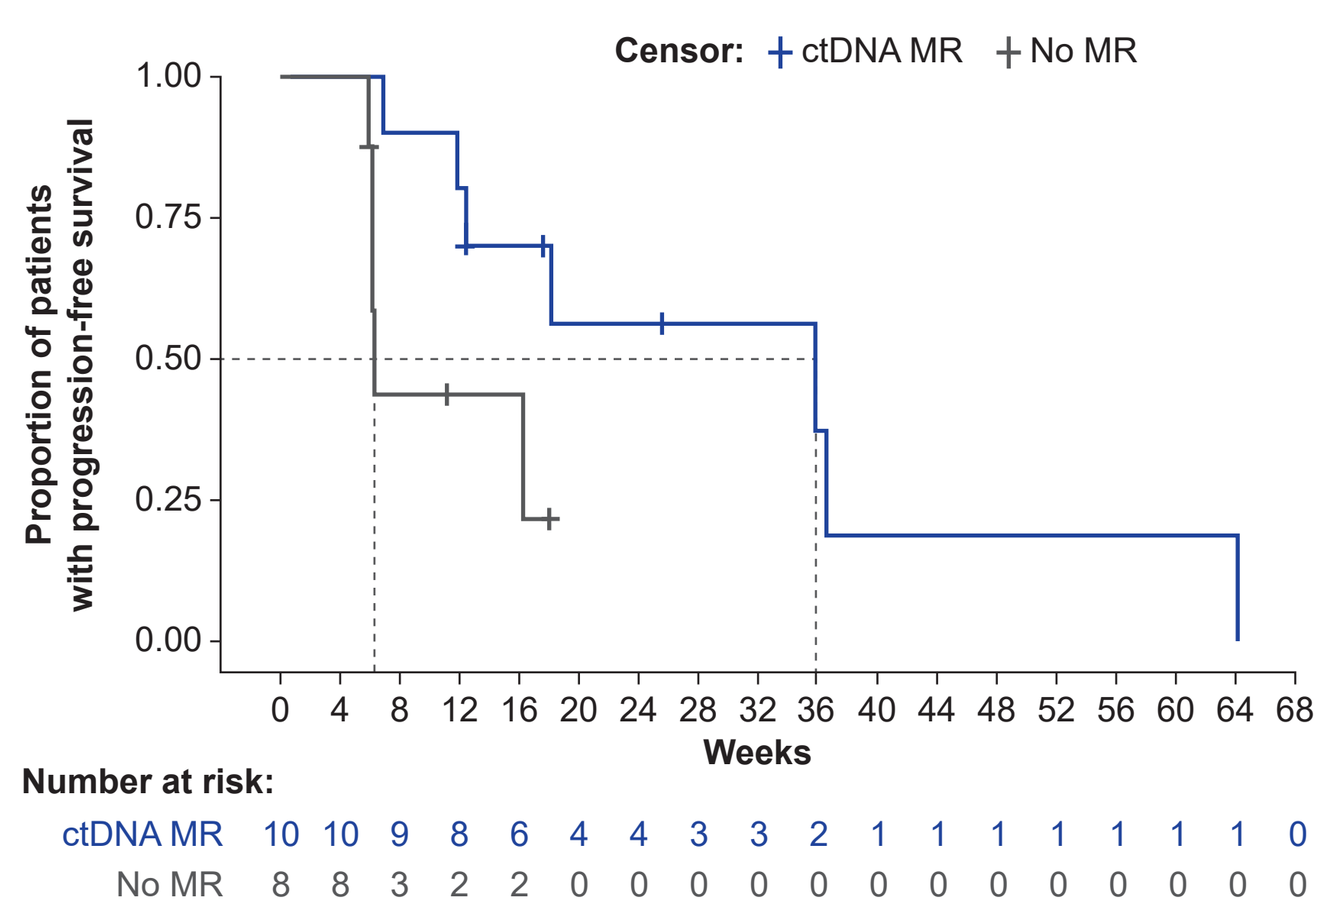
Censor: ctDNA MR No MR
Proportion of patients with progression-free survival
Weeks
Number at risk:
ctDNA MR 10 10 9 8 6 4 4 3 3 2 1 1 1 1 1 1 1 0
No MR 8 8 3 2 2 0 0 0 0 0 0 0 0 0 0 0 0 0

Figure 5: Best post-treatment reduction in ctDNA fraction

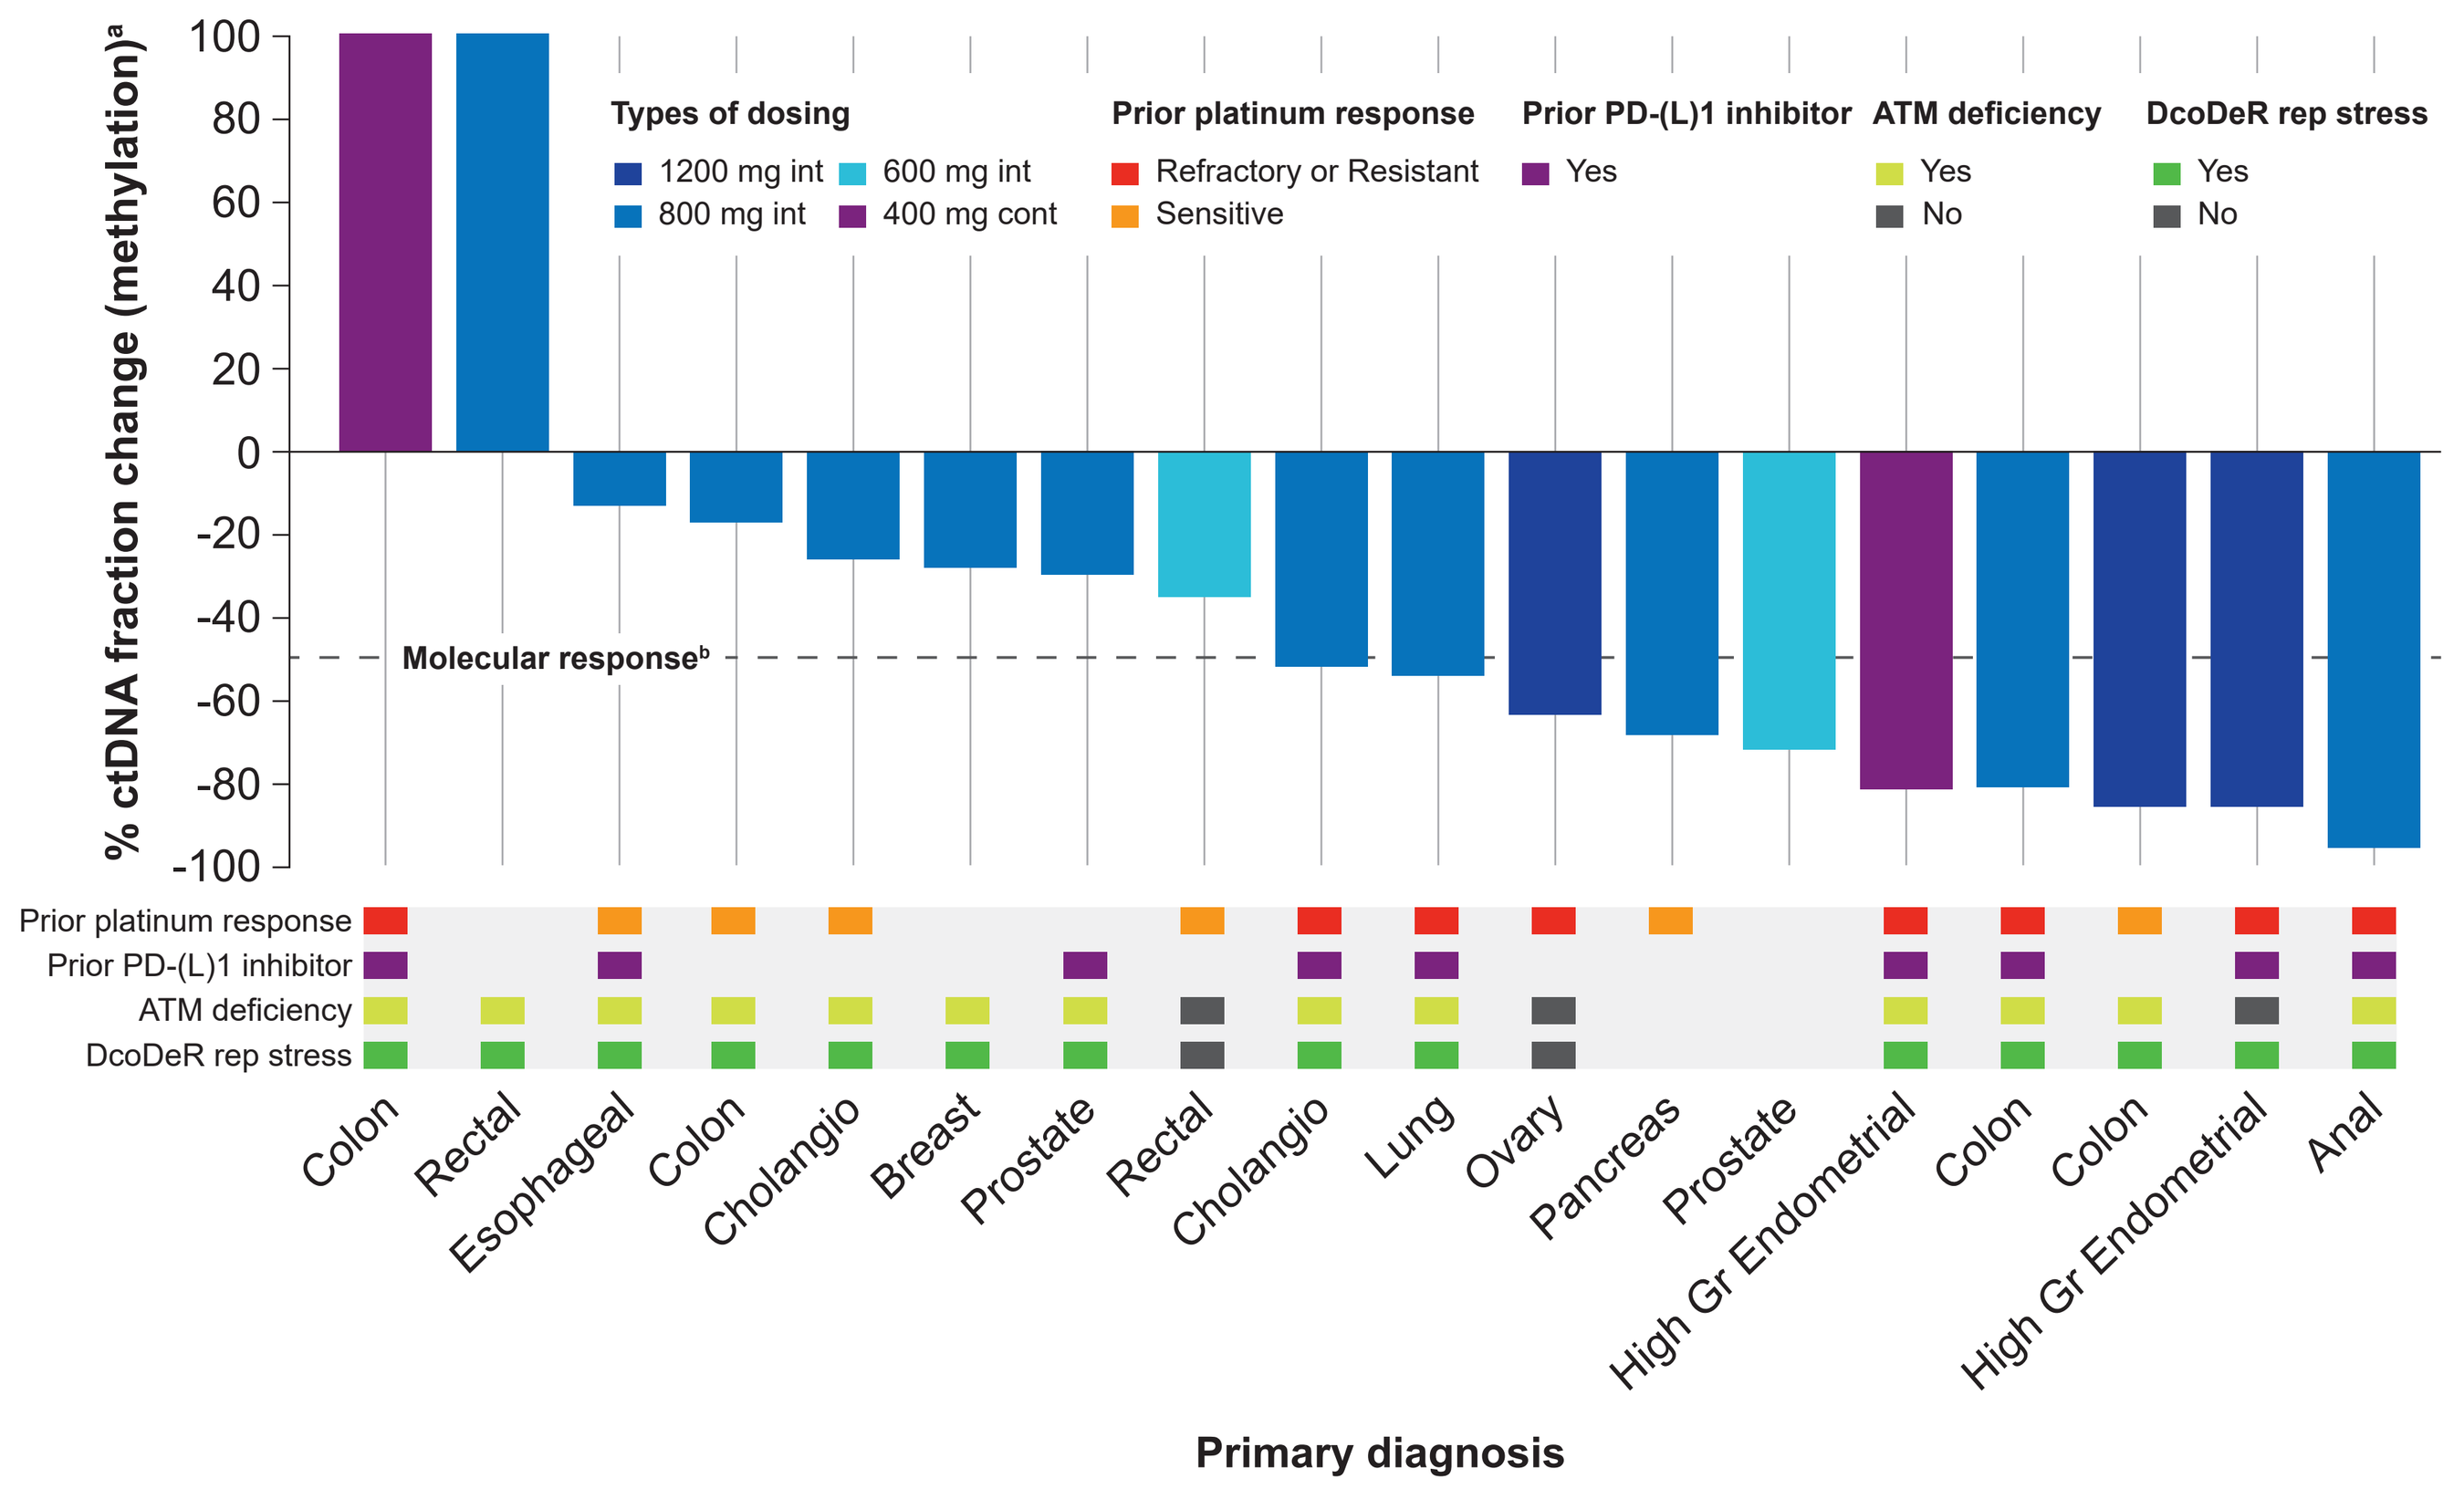
% ctDNA fraction change (methylation)[a]
Molecular response[b]
Types of dosing
1200 mg int
800 mg int
600 mg int
400 mg cont
Prior platinum response
Refractory or Resistant
Sensitive
Prior PD-(L)1 inhibitor
Yes
ATM deficiency
Yes
No
DcoDeR rep stress
Yes
No
Prior platinum response
Prior PD-(L)1 inhibitor
ATM deficiency
DcoDeR rep stress
Colon
Rectal
Esophageal
Colon
Cholangio
Breast
Prostate
Rectal
Cholangio
Lung
Ovary
Pancreas
Prostate
High Gr Endometrial
Colon
Colon
High Gr Endometrial
Anal
Primary diagnosis

[a]ctDNA analysis performed using Guardant Infinity panel; [b]Molecular response defined by ≥50% decrease in methylation-based ctDNA fraction.

Table 2: Treatment related adverse events and dose modifications

	All TRAE (≥10%) – all monotherapy doses (N = 49) Gr3 n (%)	Gr4 n (%)	All Gr n (%)	TRAE (≥10%) at doses considered tolerable (N = 22) Gr3 n (%)	Gr4 n (%)	All Gr n (%)
Any TRAE	27 (55)	2 (4)	41 (84)	8 (36)	0	17 (77)
Anemia	23 (47)	0	32 (65)	8 (36)	0	11 (50)
Fatigue	1 (2)	0	17 (35)	0	0	8 (36)
Neutropenia	8 (16)	1 (2)[a]	11 (22)	0	0	0
Nausea	0	0	9 (18)	0	0	2 (9)
Thrombocytopenia	0	1 (2)[a]	5 (10)	0	0	0
Dizziness	0	0	5 (10)	0	0	1 (5)

	Int dosing (N = 39) n (%)	Cont dosing (N = 10) n (%)
Dose interruption due to AE	24 (62)	5 (50)
Dose reduction due to AE	12 (31)	5 (50)
% planned doses omitted due to AE	14%	11%

[a]Occurred at non-tolerated dose level.

Figure 6: Best percentage change from baseline in target lesion diameter per RECIST in the efficacy analysis set

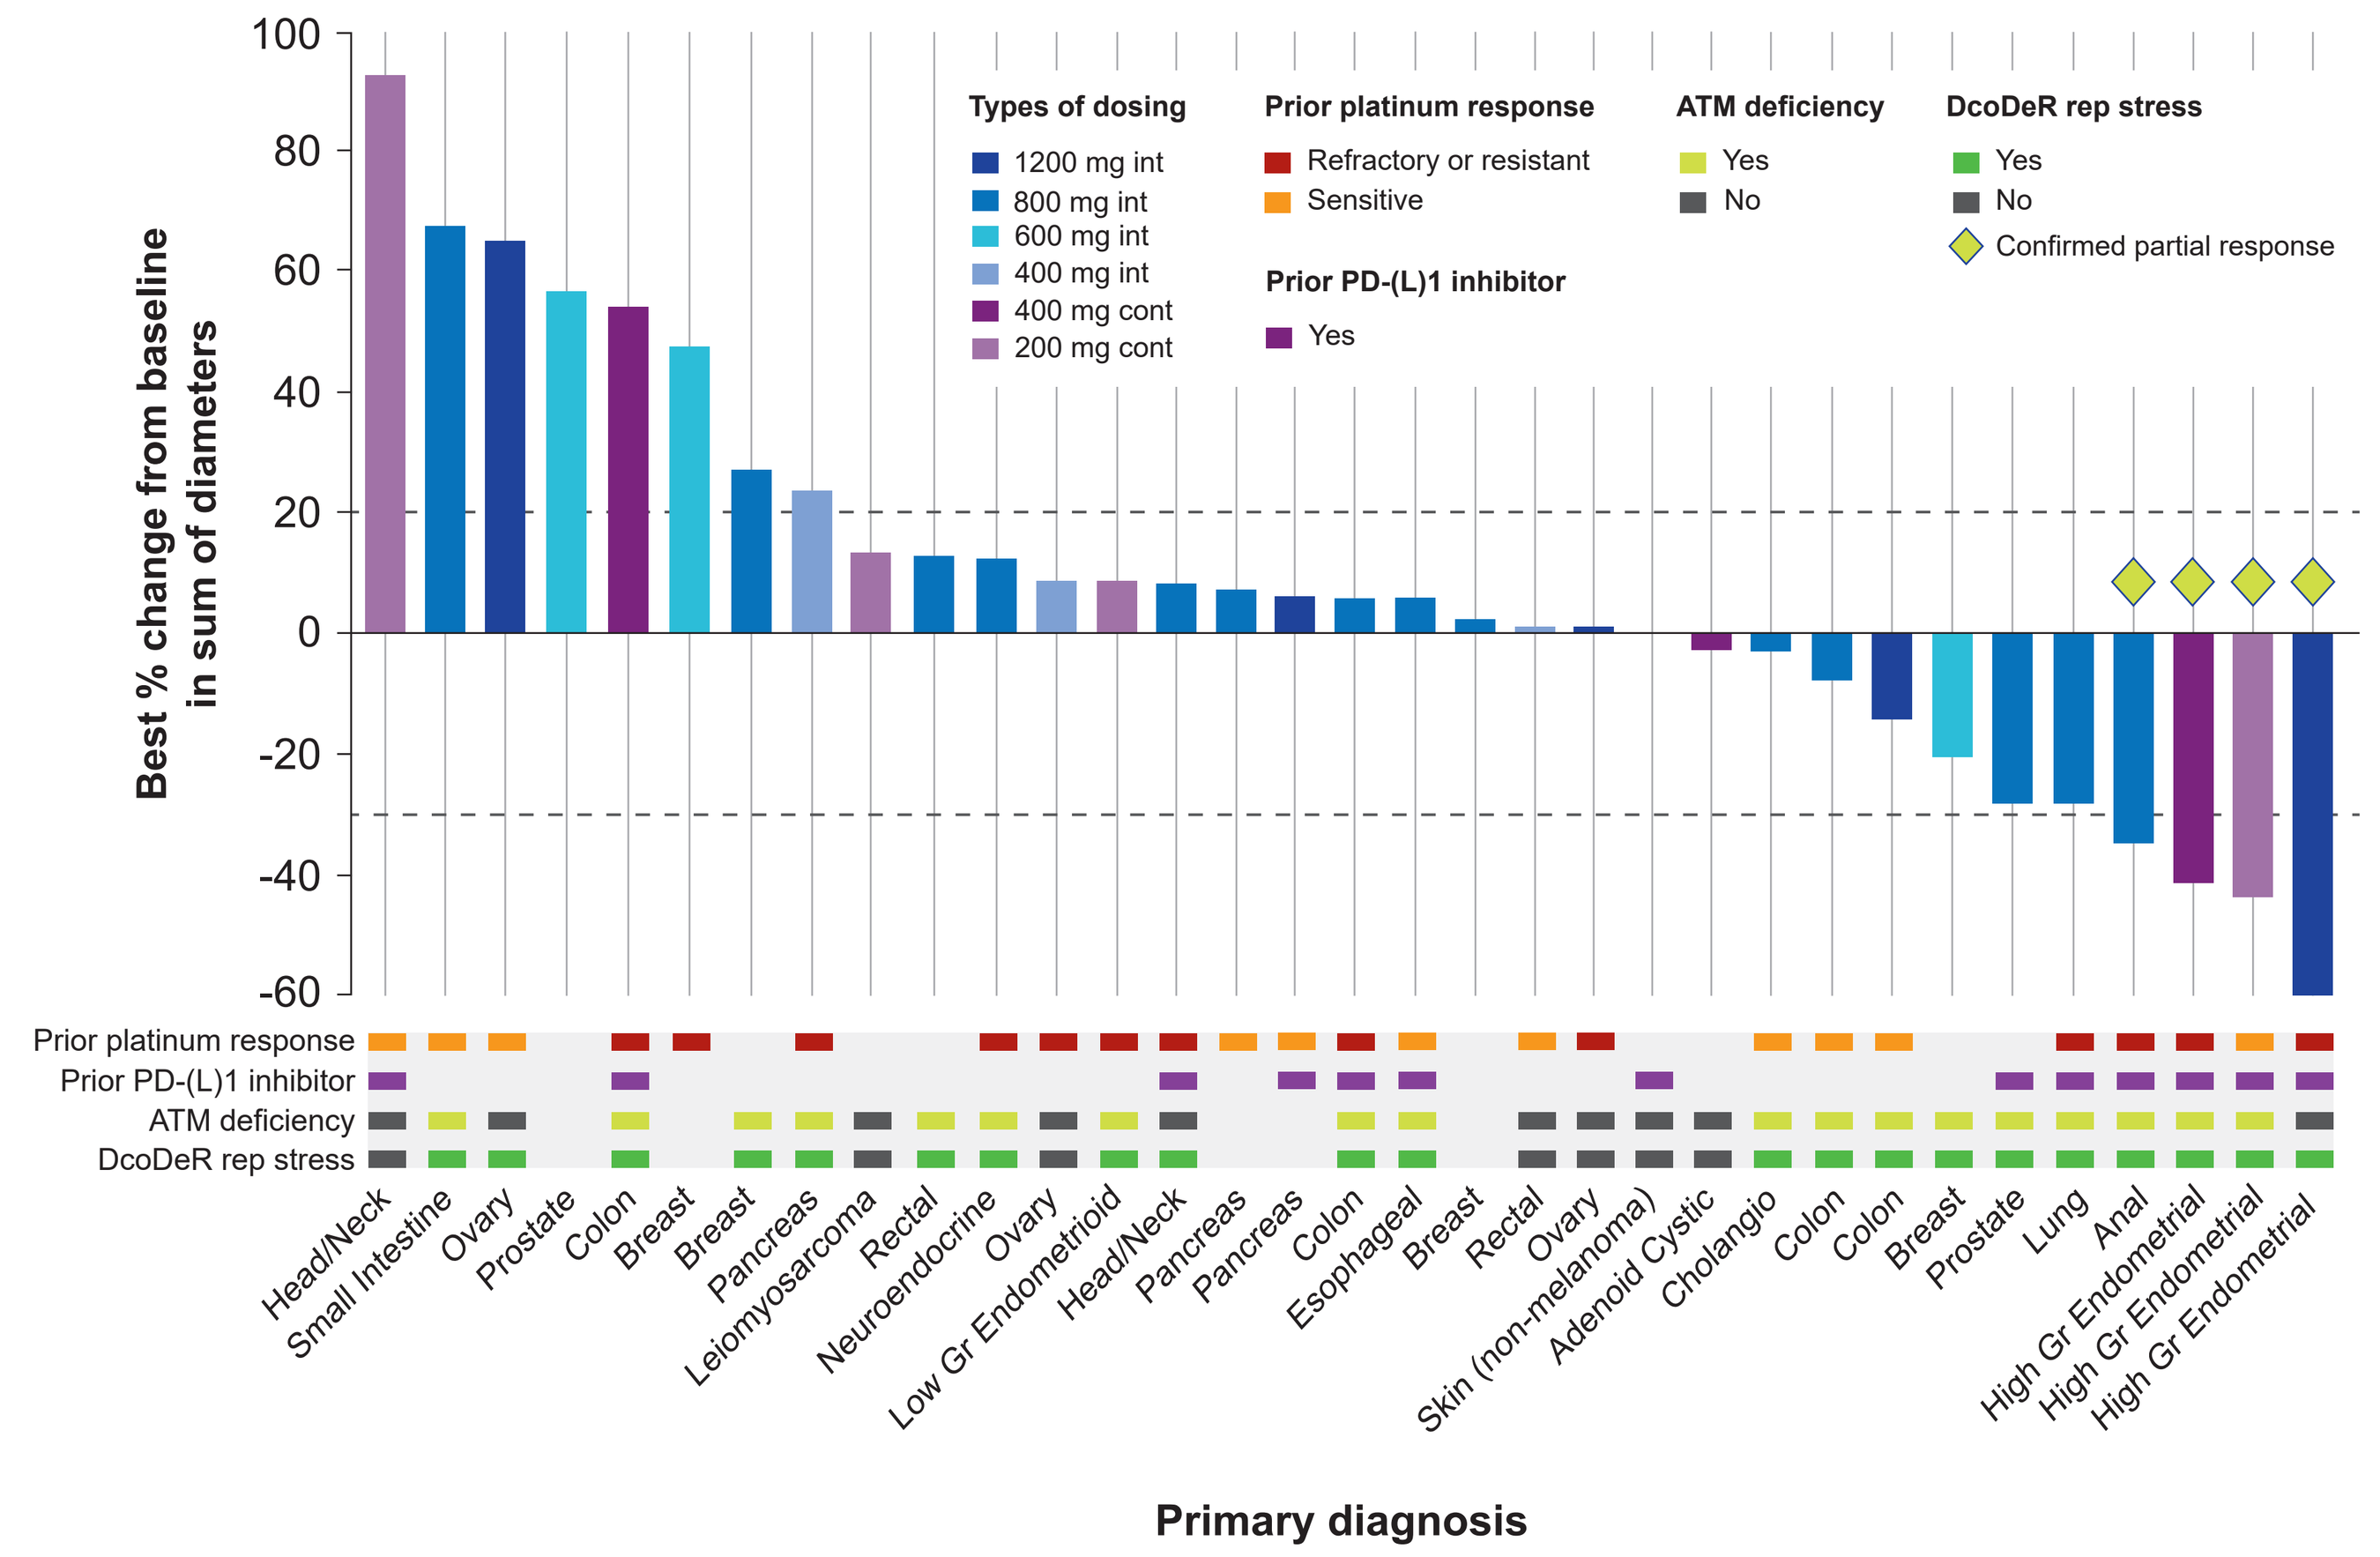
Best % change from baseline in sum of diameters
Types of dosing
1200 mg int
800 mg int
600 mg int
400 mg int
400 mg cont
200 mg cont
Prior platinum response
Refractory or resistant
Sensitive
Prior PD-(L)1 inhibitor
Yes
ATM deficiency
Yes
No
DcoDeR rep stress
Yes
No
Confirmed partial response
Prior platinum response
Prior PD-(L)1 inhibitor
ATM deficiency
DcoDeR rep stress
Head/Neck
Small Intestine
Ovary
Prostate
Colon
Breast
Breast
Pancreas
Leiomyosarcoma
Rectal
Neuroendocrine
Ovary
Low Gr Endometrioid
Head/Neck
Pancreas
Pancreas
Colon
Esophageal
Breast
Rectal
Ovary
Skin (non-melanoma)
Adenoid Cystic
Cholangio
Colon
Colon
Breast
Prostate
Lung
Anal
High Gr Endometrial
High Gr Endometrial
High Gr Endometrial
Primary diagnosis

Table 3: Characteristics of patients who responded to treatment

Age (years) Gender	68 Female	73 Female	68 Female	60 Female	63 Female
Diagnosis histology	High grade endometrial cancer	High grade endometrial cancer	High grade endometrial cancer	Anus Squamous Cell Carcinoma	Cholangiocarcinoma
Treatment history	Surgery; Paclitaxel + Carboplatin; Pembrolizumab + Lenvatinib	Surgery; Cisplatin; RT; Paclitaxel + Carboplatin + RT; Dostarlimab + Nivolumab	Surgery; Paclitaxel + Carboplatin; Doxil; Topotecan + Tamoxifen; Everolimus + Letrozole; Pembrolizumab + Lenvima; bevacizumab + olaparib + paclitaxel	RT + Mitomycin + Capecitabine; Paclitaxel + Carboplatin; Pembrolizumab; Folfox	Cisplatin + Gemcitabine + Durvalumab
Max change in tumor size (%)	-60%	-44%	-41%	-35%	Reduction in tumor size in non-measurable lesions and -92% reduction in CA19-9
Max change in ctDNA (%)	-86%	Not evaluable	-82%	-96%	-53%
Tumor biology	**ATM IHC** H score = 145, 70% positive cells **Genetic alterations:** ERBB3, FBXW7, PIK3CA, PPP2R1A, TP53, MSS	**ATM IHC** H score = 0 **Genetic alterations:** ATM, BRCA2, NF1, MLL2, ARID1A, FBXW7, RNF43, PIK3R1, PIK3R2, SETD2, SMAD4D, SPEN, MSI-H	**ATM IHC** H score = 130, 70% positive cells **Genetic alterations:** ATM, TP53, PIK3CA	**ATM IHC** H score = 17, 8% positive cells **Genetic alterations:** ATM, PIK3CA, BCL-2 amplification, MSS	**ATM IHC** H score = 0 **Genetic alterations:** ATM, MSS

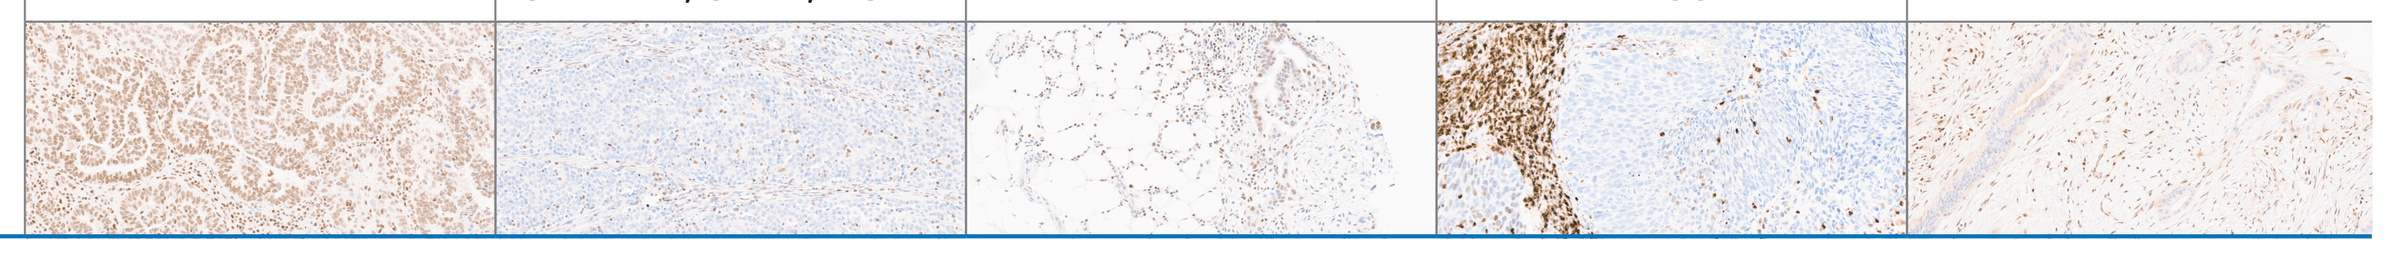

Pharmacodynamics and efficacy

- ATR-mediated DNA damage was demonstrated by an increase in γH2Ax expression in CTCs in 5/10 (50%) of evaluable post-dose CTC samples compared to pre-dose samples (Figure 3).
 - In normal cells (PBMCs) isolated from the same blood sample there was no observed trend in γH2Ax levels increasing.
- A molecular response may be associated with longer PFS in patients treated with ART0380 (Figure 4).
 - ART0380 treatment was associated with molecular response in a variety of cancers with genotypes that may be indicative of higher endogenous replication stress (Figure 5).
- Reduction in tumor size was observed in patients with cancers of multiple histologies (Figure 6); many of these had molecular alterations reported to cause replication stress.
 - All 3 patients with high grade endometrial cancer enrolled experienced durable confirmed responses.

Figure 7: Treatment duration in the efficacy analysis set

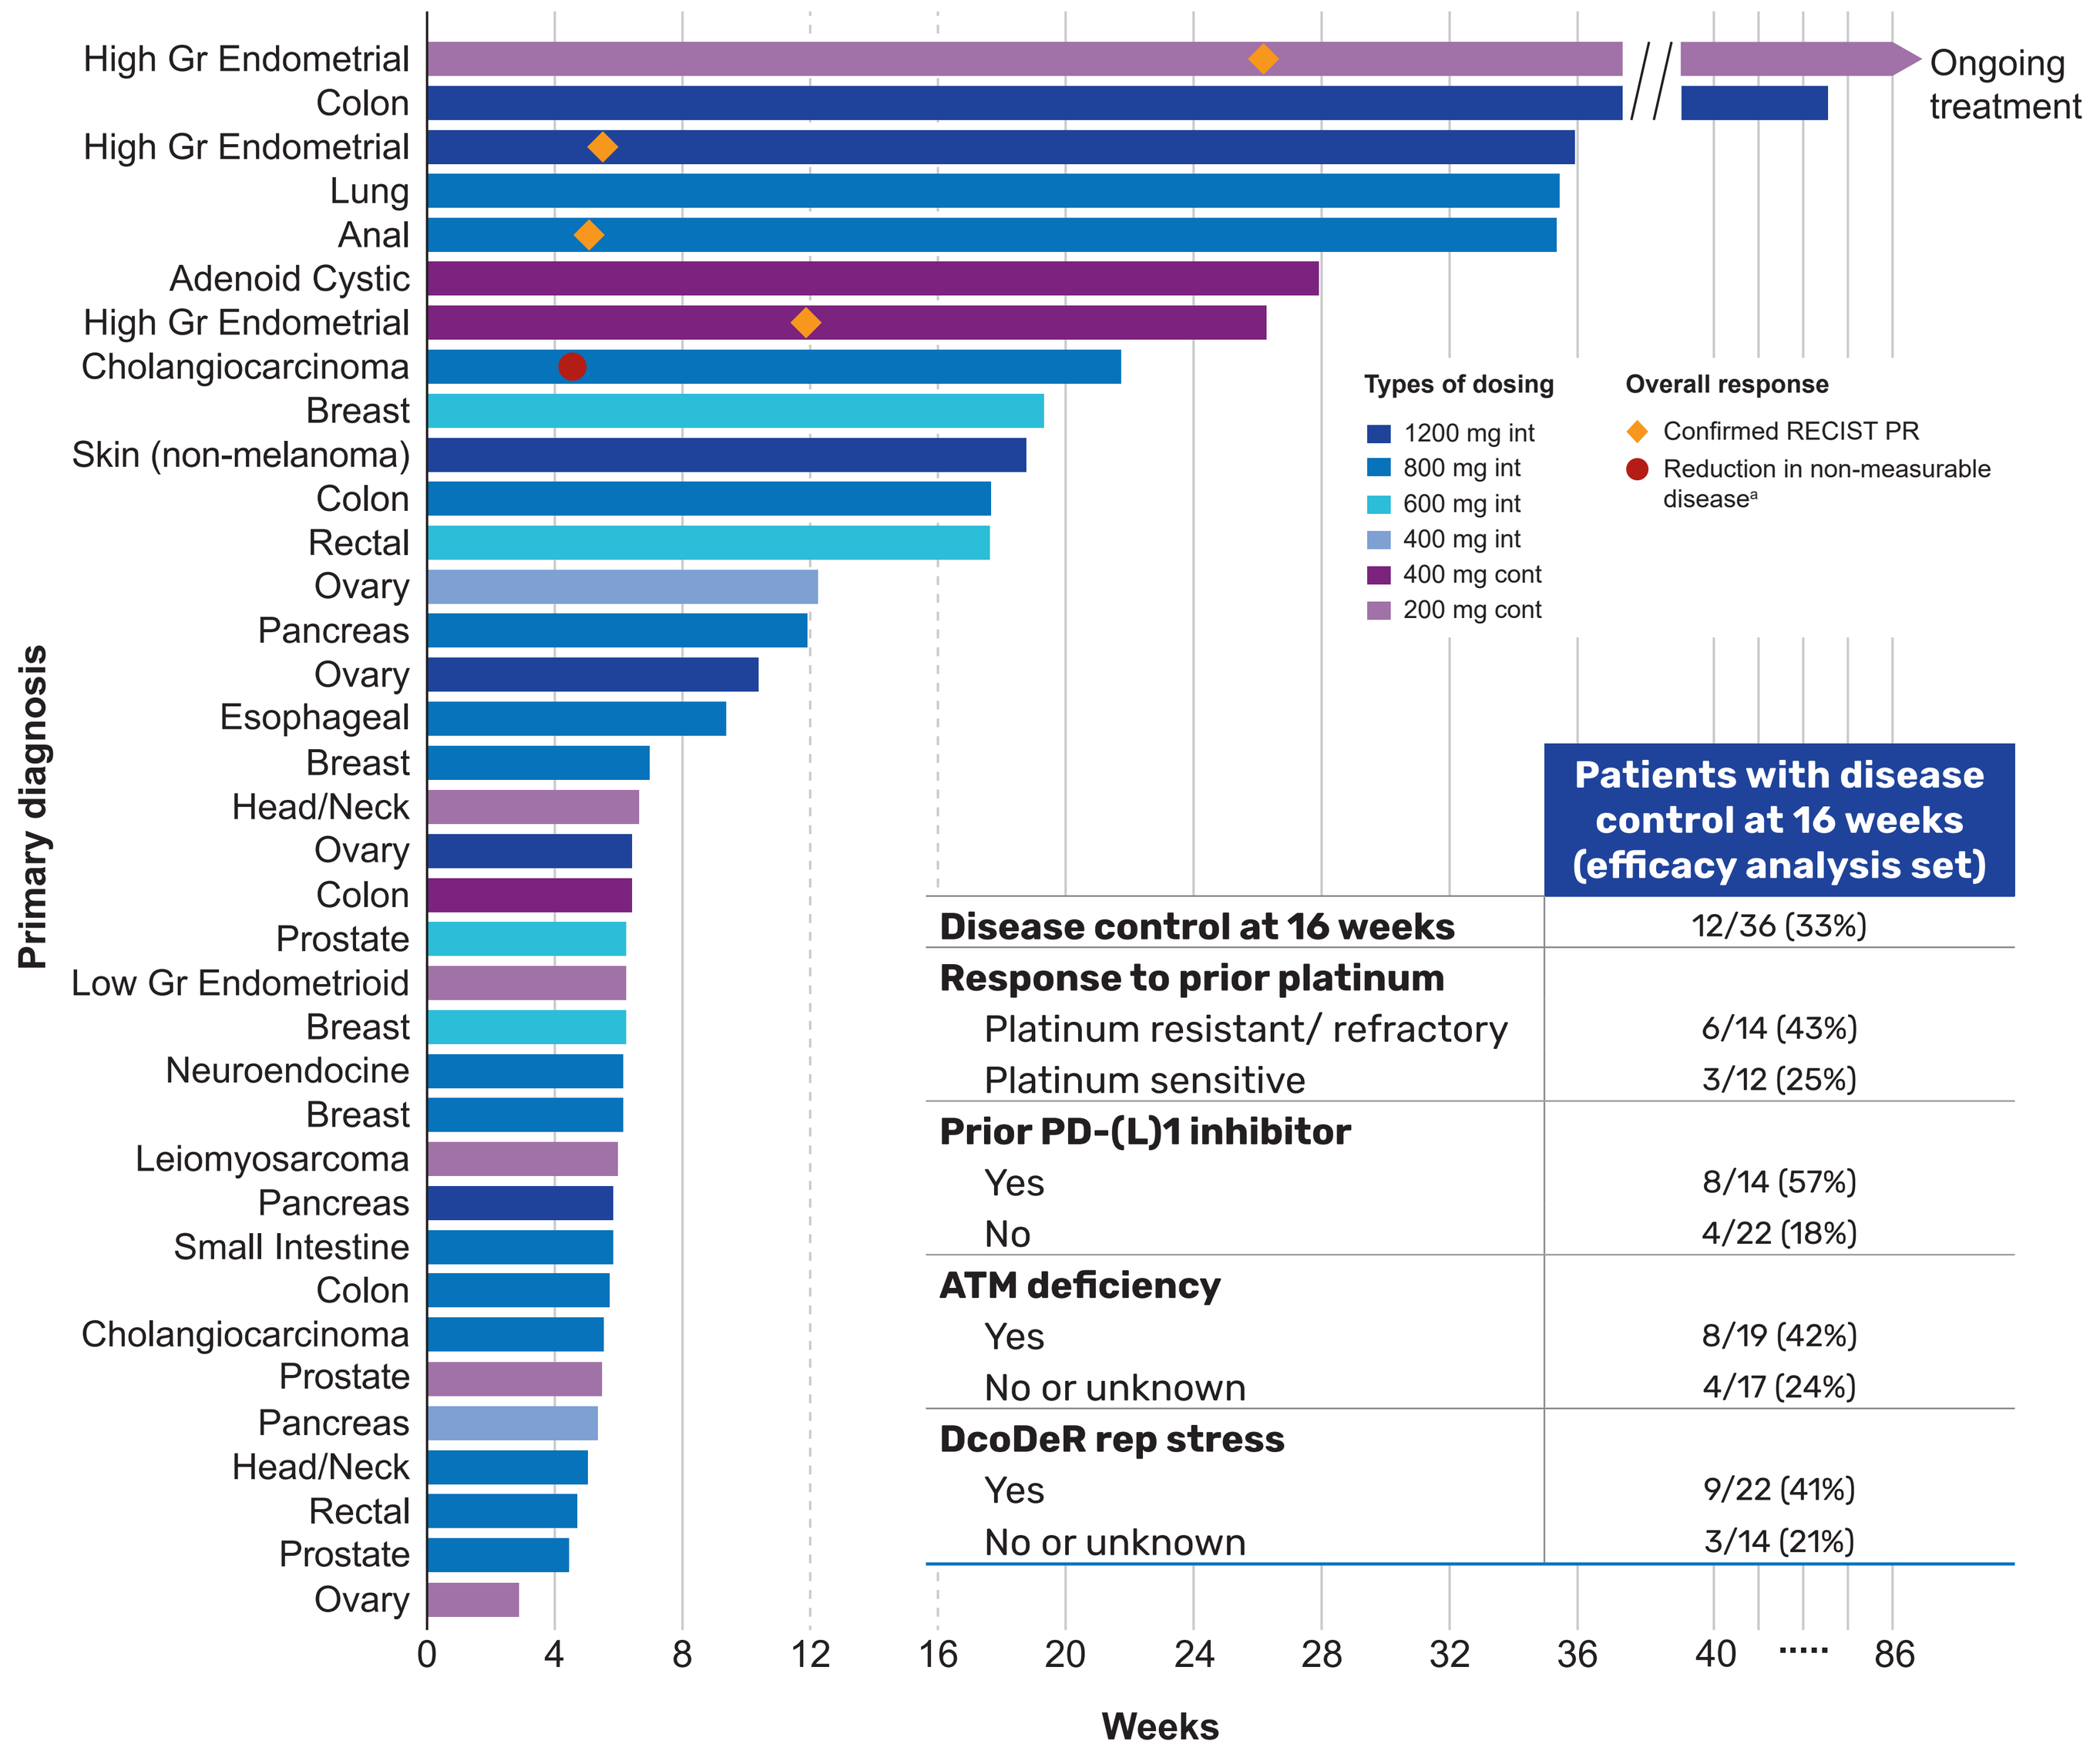
High Gr Endometrial
Colon
High Gr Endometrial
Lung
Anal
Adenoid Cystic
High Gr Endometrial
Cholangiocarcinoma
Breast
Skin (non-melanoma)
Colon
Rectal
Ovary
Pancreas
Ovary
Esophageal
Breast
Head/Neck
Ovary
Colon
Prostate
Low Gr Endometrioid
Breast
Neuroendocine
Breast
Leiomyosarcoma
Pancreas
Small Intestine
Colon
Cholangiocarcinoma
Prostate
Pancreas
Head/Neck
Rectal
Prostate
Ovary
Primary diagnosis
Ongoing treatment
Types of dosing
1200 mg int
800 mg int
600 mg int
400 mg int
400 mg cont
200 mg cont
Overall response
Confirmed RECIST PR
Reduction in non-measurable disease[a]
0
4
8
12
16
20
24
28
32
36
40
86
Weeks

	Patients with disease control at 16 weeks (efficacy analysis set)
Disease control at 16 weeks	12/36 (33%)
Response to prior platinum	
Platinum resistant/ refractory	6/14 (43%)
Platinum sensitive	3/12 (25%)
Prior PD-(L)1 inhibitor	
Yes	8/14 (57%)
No	4/22 (18%)
ATM deficiency	
Yes	8/19 (42%)
No or unknown	4/17 (24%)
DcoDeR rep stress	
Yes	9/22 (41%)
No or unknown	3/14 (21%)

[a]With >50% decline in tumor marker.

CONCLUSIONS

- ART0380 is safe and well tolerated when dosed intermittently up to 600 mg and at 200 mg continuously.
 - At these doses, the on-target anemia was predictable, manageable, and reversible.
- ART0380 demonstrated a PK profile with rapid absorption followed by relatively rapid elimination with dose proportional exposure which was of low variability.
- ART0380 induced DNA damage as shown by increased γH2Ax in CTCs from patients whose tumor harbored a DDR deficiency. There was no trend of increasing γH2Ax in normal cells (PBMCs) in these patients.
- ART0380 was shown to be clinically effective with patients who received ART0380 experiencing both molecular and clinical responses.
 - There was a trend for patients who experienced a molecular response to have a longer progression free survival
 - Five patients with tumors that exhibited biology consistent with higher replication stress experienced clinical responses.
- Clinical evaluation of ART0380 as monotherapy and in combination with chemotherapy in patients with cancers of selected biology is ongoing.

Abbreviations: ATR: Ataxia telangiectasia and Rad3-related; **ATM :** ataxia-telangiectasia-mutated; **AUC:** area under the curve; **Cholangio:** Cholangiocarcinoma; **CTC:** circulating tumor cells; **ctDNA:** circulating tumor DNA; **DDR:** DNA damage response; **DLT:** dose-limiting toxicity; **DNA:** deoxyribonucleic acid; **EC_{50}:** half maximal effective concentration; **ECOG:** Eastern Cooperative Oncology Group; **Gr:** Grade; **IHC:** immunohistochemistry; **Leiomyo:** Leiomyosarcoma; **PARP:** poly (ADP-ribose) polymerase; **PBMC:** peripheral blood mononuclear cells; **PD-(L)1:** programmed death ligand 1; **PFS:** progression free survival; **RP2D:** monotherapy recommended phase 2 dose; **TRAE:** treatment related adverse event; **γH2Ax:** gamma H2Ax.

References: 1. Majithiya et al. Cancer Res 2023;83(7_Suppl):Abstract 312. **Presenting author disclosures:** Participated in advisory boards for: Aadi, AstraZeneca, Aravive, Alkemeres, Blueprint Pharma, Caris, Clovis, Duality, Eisai, Eli Lilly, EMD Serono, GlaxoSmithKline/Tesaro, Genetech/Roche, Hengrui, Immunogen, IMab, Merck, Mereo, Myriad, Mersana, Novartis, Novocure, OncXerna, OncoNova, Pannavance, Tarveda, VBL Therapeutics, Verastem, Zentalis; Associate director of: GOG Partners; Chair of: NRG Ovarian Cancer; Member of the board of directors for: ASCO, GOG Foundation; Royalties from: UP to Date; Prinicpal Investigator for: Artios, Astra Zeneca, Bolt, Daiichi Sankyo, Duality, Eli Lilly, GlaxoSmithKline/Tesaro, Immunogen, PTC Therapeutics, Regeneron, Verastem. **Acknowledgements:** We would like to thank the patients, their families, study sites and study personnel who have participated in the study. We would also like to thank the Costello Medical Creative Team for design support, and Erin Clarkson, BSc, Costello Medical, for editorial assistance; Guardant Health for performing ctDNA analysis and Precision for Medicine for performing the CTC analysis. This study was funded by Artios. All costs associated with development of this poster were funded by Artios. ART0380 was identified through a collaboration between MDACC and ShangPharma. The compound was in-licensed by Artios in 2019.

Presented at ESMO 2023 | Madrid, Spain | 20–24 October 2023

This presentation is the intellectual property of Artios Pharma Ltd. Contact the presenting author at Kathleen-Moore@ouhsc.edu for permission to reprint and/or distribute. Sponsored by Artios Pharma Ltd.